Novel Target Identification Using Oxford BioTherapeutics' Proprietary Proteomics Target Discovery Platform, OGAP®, for First-in-Class ADCs and Other Antibody Therapeutics.

Poster Number 13

Somdatta Basu, Abigail Houghton, Benjamin Thomas, Lindsey Hudson, Daniel Davies, Joseph Lewis,
Jason Allen, Martin Barnes, Stephen Blance and Christian Rohlff
Oxford BioTherapeutics Ltd, 94a Innovation Drive, Milton Park, Abingdon, OX14 4RZ, UK
www.oxfordbiotherapeutics.com
Contact: Somdatta.B@oxfordbiotherapeutics.com

Abstract

Oxford BioTherapeutics (OBT) is a clinical stage oncology company specialising in identifying novel cancer therapeutic targets using its unique, proprietary, proteomic target discovery platform, OGAP® and developing first-in-class, antibody-based therapies in: Antibody Drug Conjugate (ADC), Immuno-oncology (IO), Chimeric Antigen Receptor T cell (CAR-T), monoclonal antibody (mAb) and T-cell Engager (TCE). OGAP® is one the world's largest quantitative membrane protein expression libraries, generated using proteomics. OGAP® contains data from multiple patient tumors, normal adjacent tissues and normal tissues. OGAP® successfully predicts therapeutic indices of multiple, current, clinical-stage targets showcasing its ability to highlight toxicity/safety and efficacy of such targets by directly viewing OGAP® generated quantitative metrics. Directly measuring membrane protein abundance circumvents the problem of poor correlation of mRNA abundance with protein expression[1]. This has allowed OBT to identify new targets which have been previously missed by mRNA analysis alone, which most of the industry relies on.
OBT's internal pipeline comprises of its lead ADC phase 1 clinical asset, OBT076, an IO monotherapy and two IO bispecifics in preclinical development. OBT's successful strategy is externally validated by top-tier partnerships in oncology. OBT's externally partnered pipeline assets include, two TCEs in phase 1 clinical trials and multiple TCEs in preclinical and early development; an IO monotherapy in preclinical development and multiple ADCs in preclinical and early development.

1 Introduction

- OBT utilises its unique, proprietary, proteomic target discovery platform, OGAP® to identify novel, first-in-class, antibody-therapeutic targets.
- Target characteristics can be tailored for different drug modalities: ADC, mAb, TCE, IO and CAR-T.
- OGAP® contains data from ~650 patient tumor tissues and cell lines, covering all major liquid and solid, both primary and metastatic, tumor indications, critical normal/normal adjacent tissues (NATs), as well as cell lines.
- Data in OGAP® has been generated by directly measuring membrane protein abundance in patient tissues using quantitative mass spectrometry based proteomics.
- OBT's approach to identifying new oncology targets by quantitative proteomics overcomes the issue of poor correlation (~40%) of mRNA abundance with protein expression[1] and does not rely on the widely used public mRNA datasets (TCGA, GTex).
- OGAP® quantitative metrics predict toxicity/safety and efficacy/tolerability concerns of multiple, known, clinical-stage targets.
- OBT's unique, successful strategy is externally validated by multiple top-tier collaborators in oncology in ADCs, TCEs, CAR-Ts and IO.

2 OBT's Target Discovery Workflow

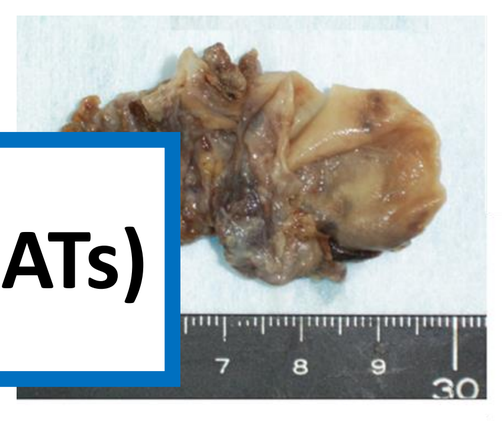
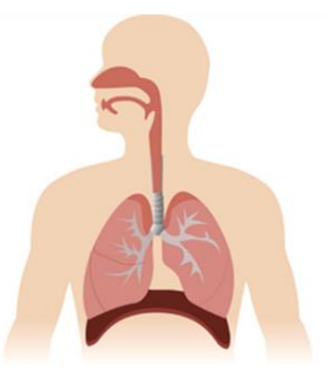

Tumor Tissues and Normal/Normal Adjacent Tissues (NATs)

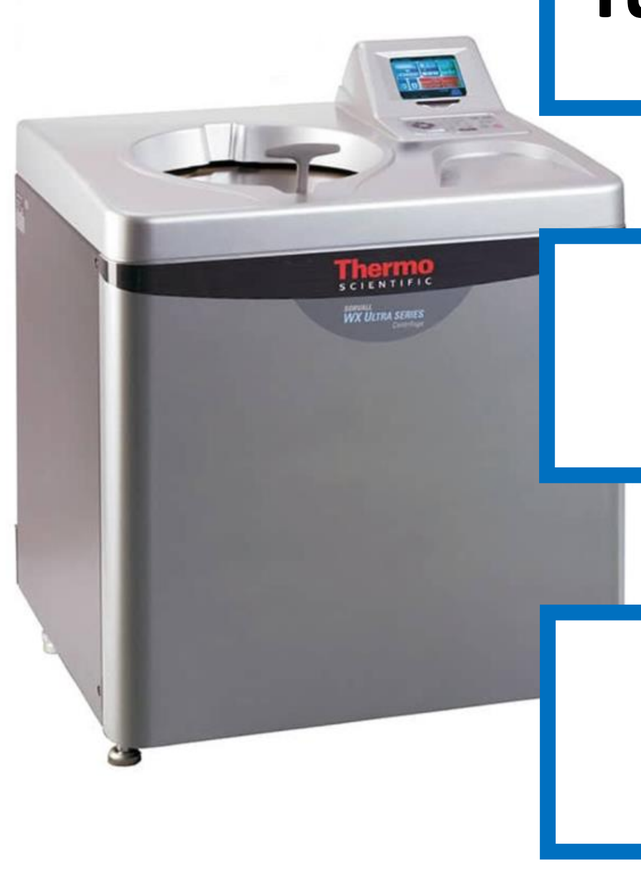

Thermo

Membrane Extraction from Tissues to Obtain Proteins

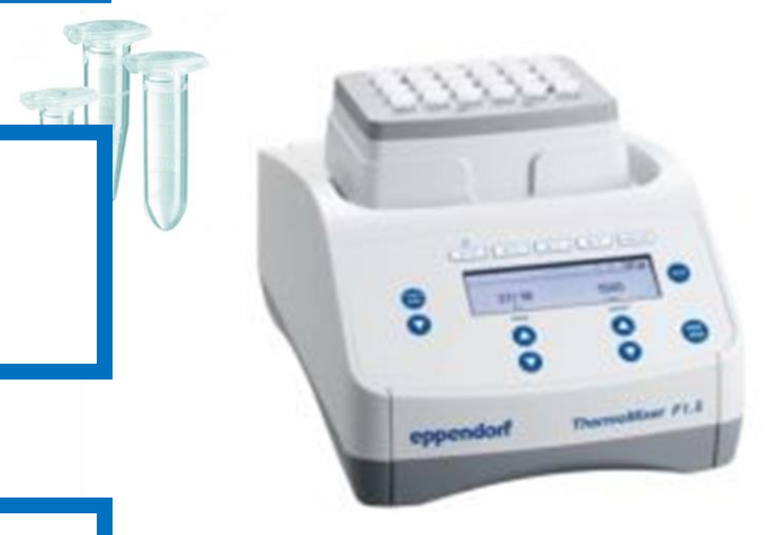

eppendorf

Tryptic Digestion of Proteins

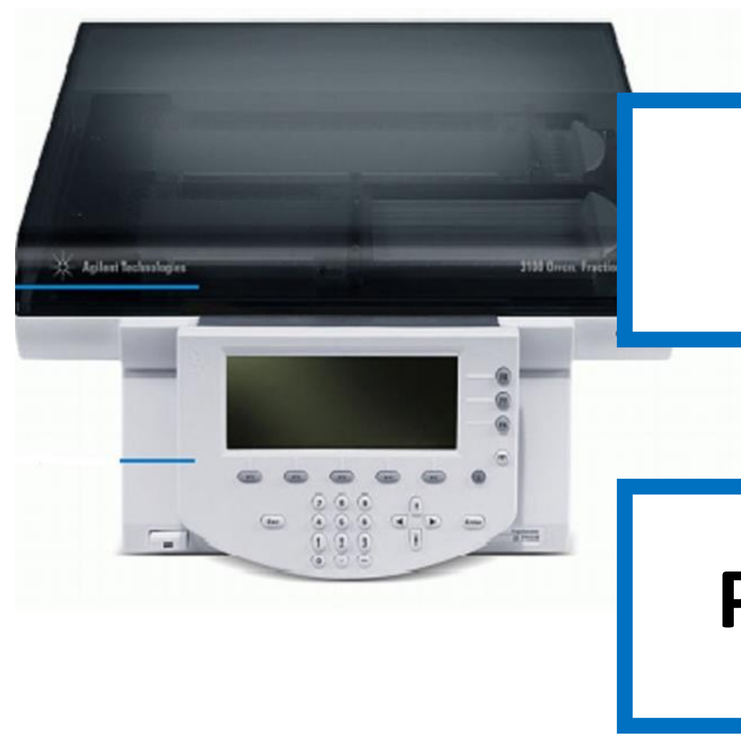

Peptides are Fractionated

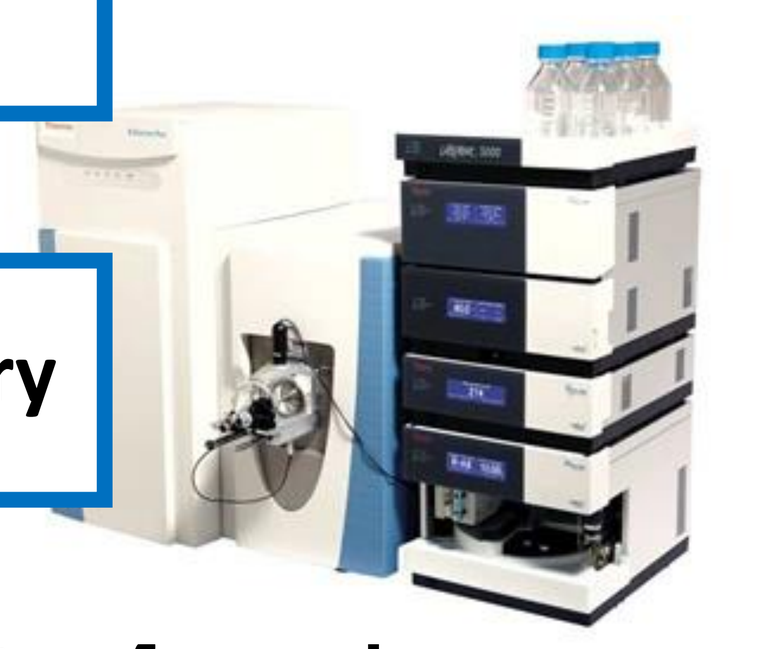

Proteins Identified and Quantified by Mass Spectrometry

OGAP contains cancer and normal tissue expression profiles of majority of membrane proteins (5000-7000 proteins)

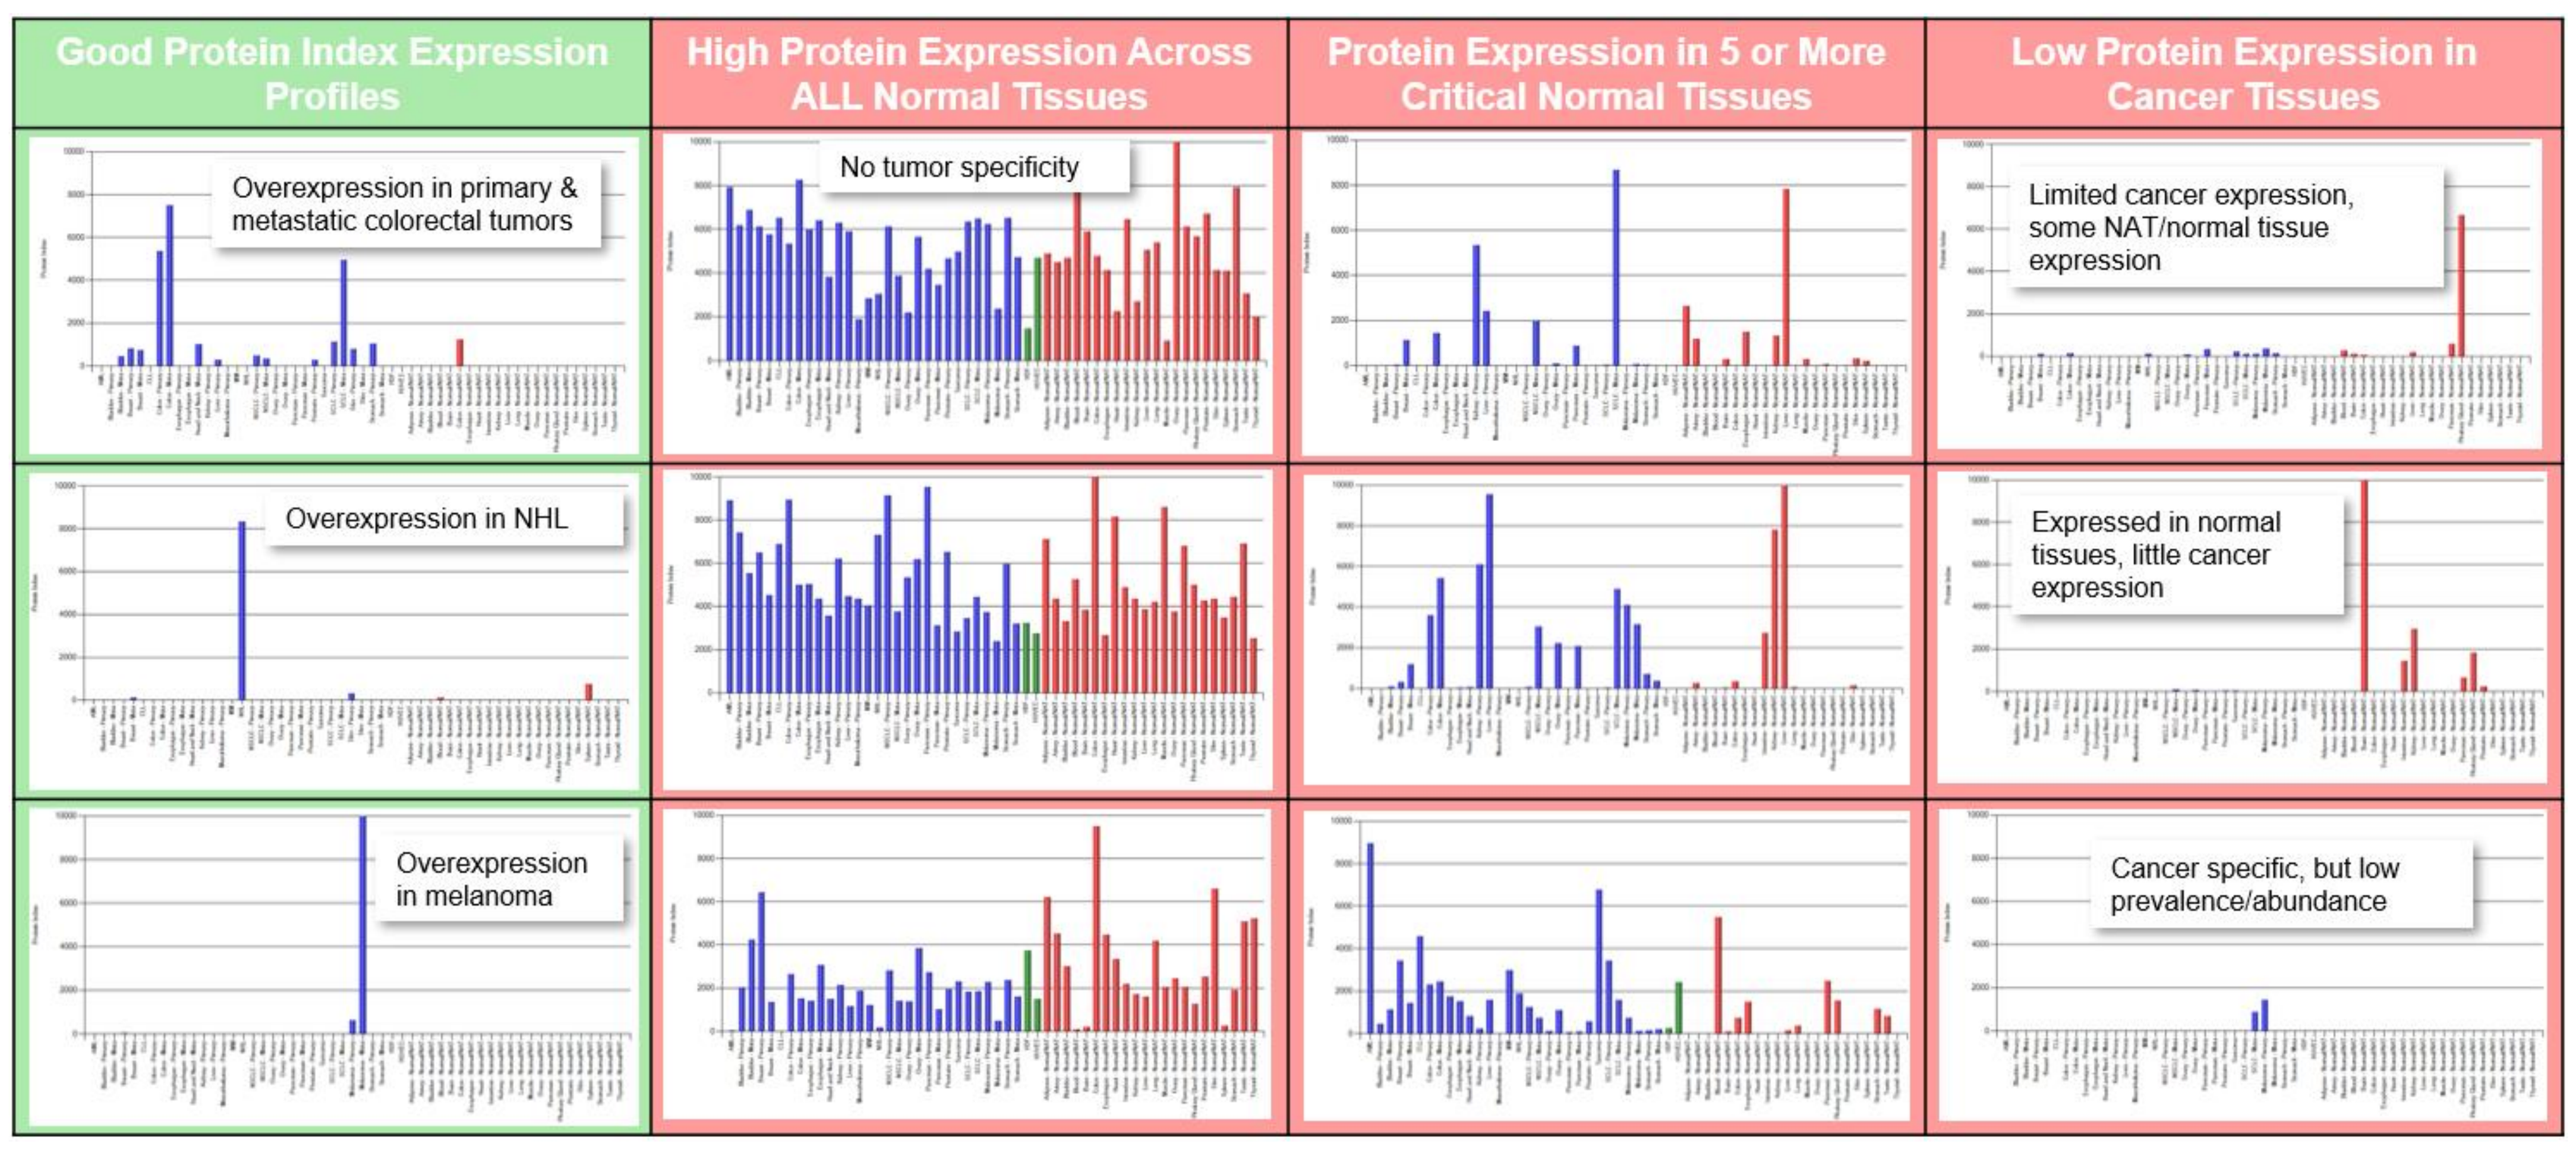

Good Protein Index Expression Profiles
High Protein Expression Across ALL Normal Tissues
Protein Expression in 5 or More Critical Normal Tissues
Low Protein Expression in Cancer Tissues
Overexpression in primary & metastatic colorectal tumors
No tumor specificity
Limited cancer expression, some NAT/normal tissue expression
Overexpression in NHL
Expressed in normal tissues, little cancer expression
Overexpression in melanoma
Cancer specific, but low prevalence/abundance

Figure 1: OGAP Protein Index Plots of Different Membrane Proteins. Showing examples of good cell surface protein expression profiles (in green box) in Colorectal Cancer, Non-Hodgkin Lymphoma and Melanoma. Several examples of membrane proteins which are not good targets, with expression in most normal tissues or limited expression in cancers (in red box) are shown.

OGAP Protein Index (PI) predicts therapeutic indices of clinical targets

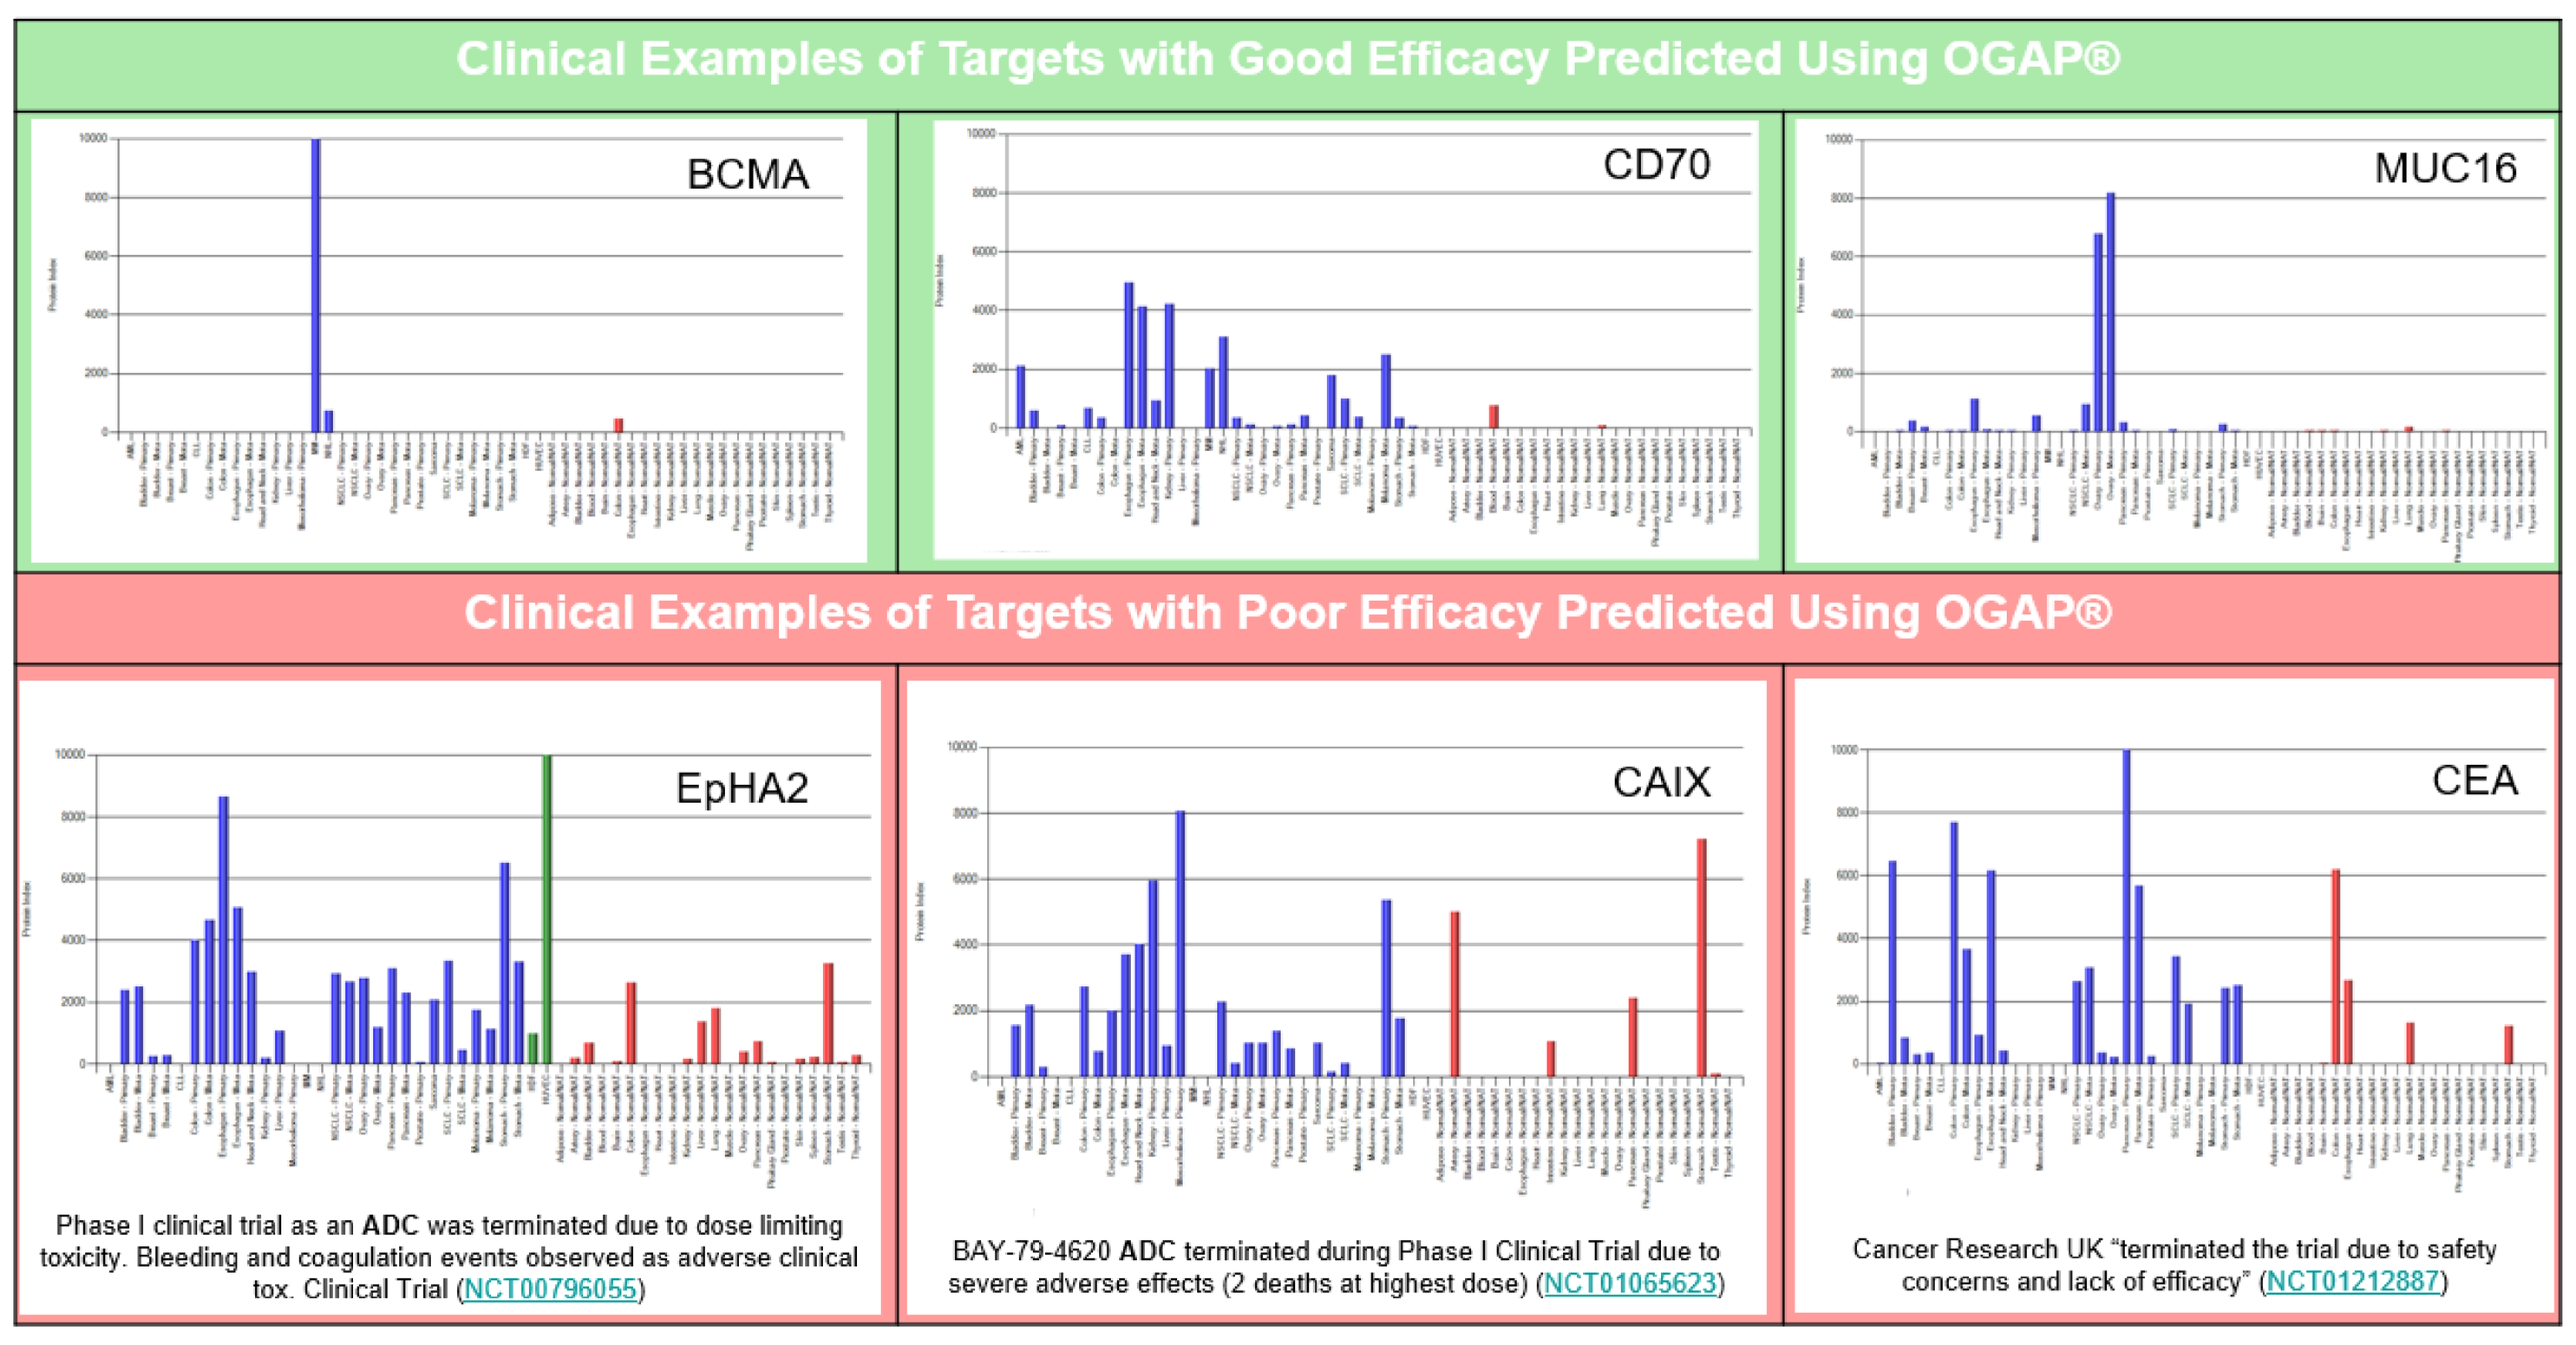

Clinical Examples of Targets with Good Efficacy Predicted Using OGAP®
BCMA
CD70
MUC16
Clinical Examples of Targets with Poor Efficacy Predicted Using OGAP®
EpHA2
CAIX
CEA
Phase I clinical trial as an ADC was terminated due to dose limiting toxicity. Bleeding and coagulation events observed as adverse clinical tox. Clinical Trial (NCT00796055)
BAY-79-4620 ADC terminated during Phase I Clinical Trial due to severe adverse effects (2 deaths at highest dose) (NCT01065623)
Cancer Research UK "terminated the trial due to safety concerns and lack of efficacy" (NCT01212887)

Figure 2: OGAP Protein Index of Known Clinical Targets. Showing examples of protein expression of clinical targets (BCMA, CD70 and MUC16) with good efficacy/tolerability (in green box) and examples of protein expression of clinical targets (EpHA2, CAIX and CEA) with poor efficacy/tolerability (in red box).

3 OGAP Data Analysis for Protein Target Selection

3.1 The Protein Index (PI) plot gives a semi-quantitative overview of the expression of an individual protein across cancer and normal tissues.

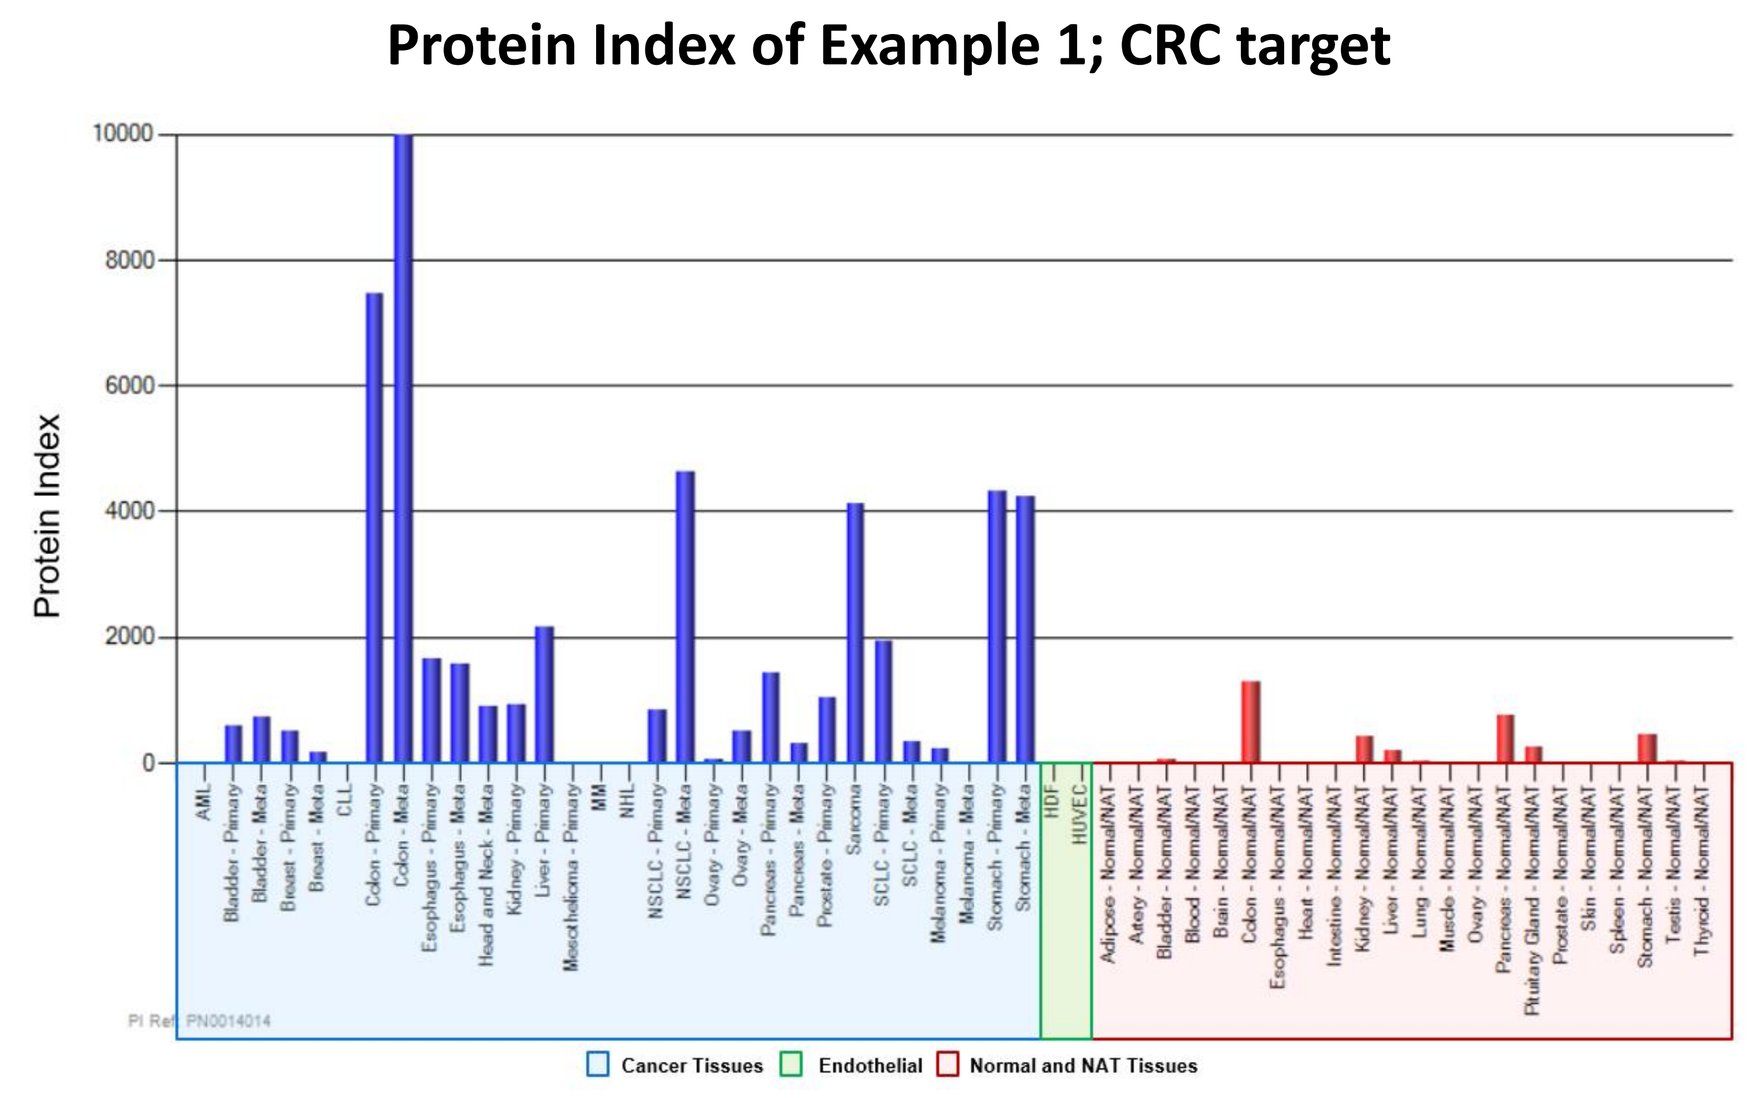

Protein Index of Example 1; CRC target
Protein Index
Cancer Tissues
Endothelial
Normal and NAT Tissues

Figure 3: Protein Index Plot for Example 1. A semi quantitative metric combining abundance and prevalence of a protein from over 650 patient and cell line samples. Cancer indications are in blue, endothelial cells in green and NAT/normal tissues in red.

3.2 Next, a more detailed drill-down of patient prevalence & target abundance in different patients is examined for both primary and metastatic diseases

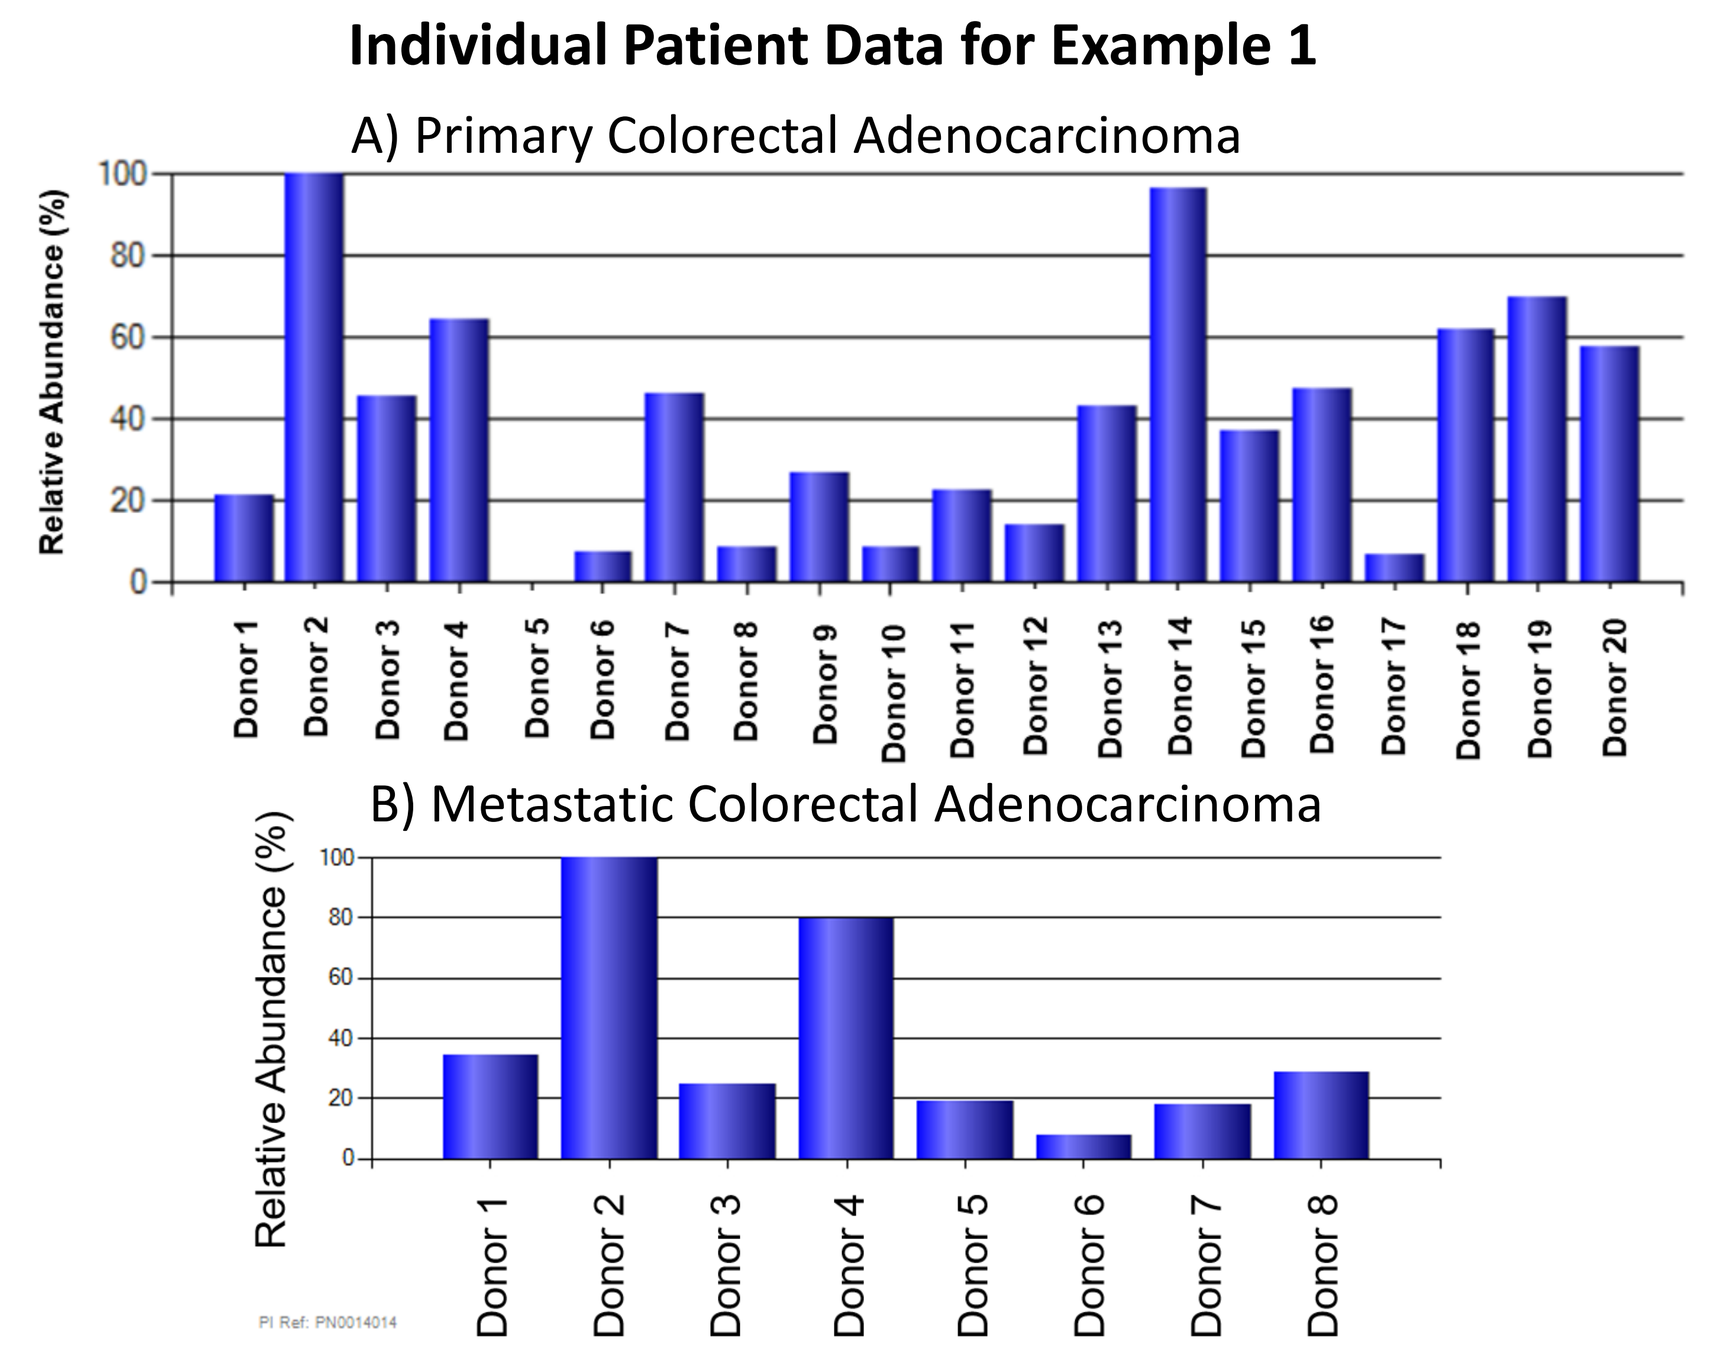

Individual Patient Data for Example 1
A) Primary Colorectal Adenocarcinoma
Relative Abundance (%)
B) Metastatic Colorectal Adenocarcinoma
Relative Abundance (%)

Figure 4: Patient Sample Breakdowns for Example 1. Showing membrane protein expression for A) primary colorectal adenocarcinoma and B) metastatic colorectal adenocarcinoma

3.3 Estimation of plasma membrane (PM) copy number of the target protein. As we measure all PM proteins within a particular indication we can estimate the cell surface abundance of a target protein, relative to all cell surface proteins in that indication. Copy number can be estimated from the plot relative to known benchmarks.

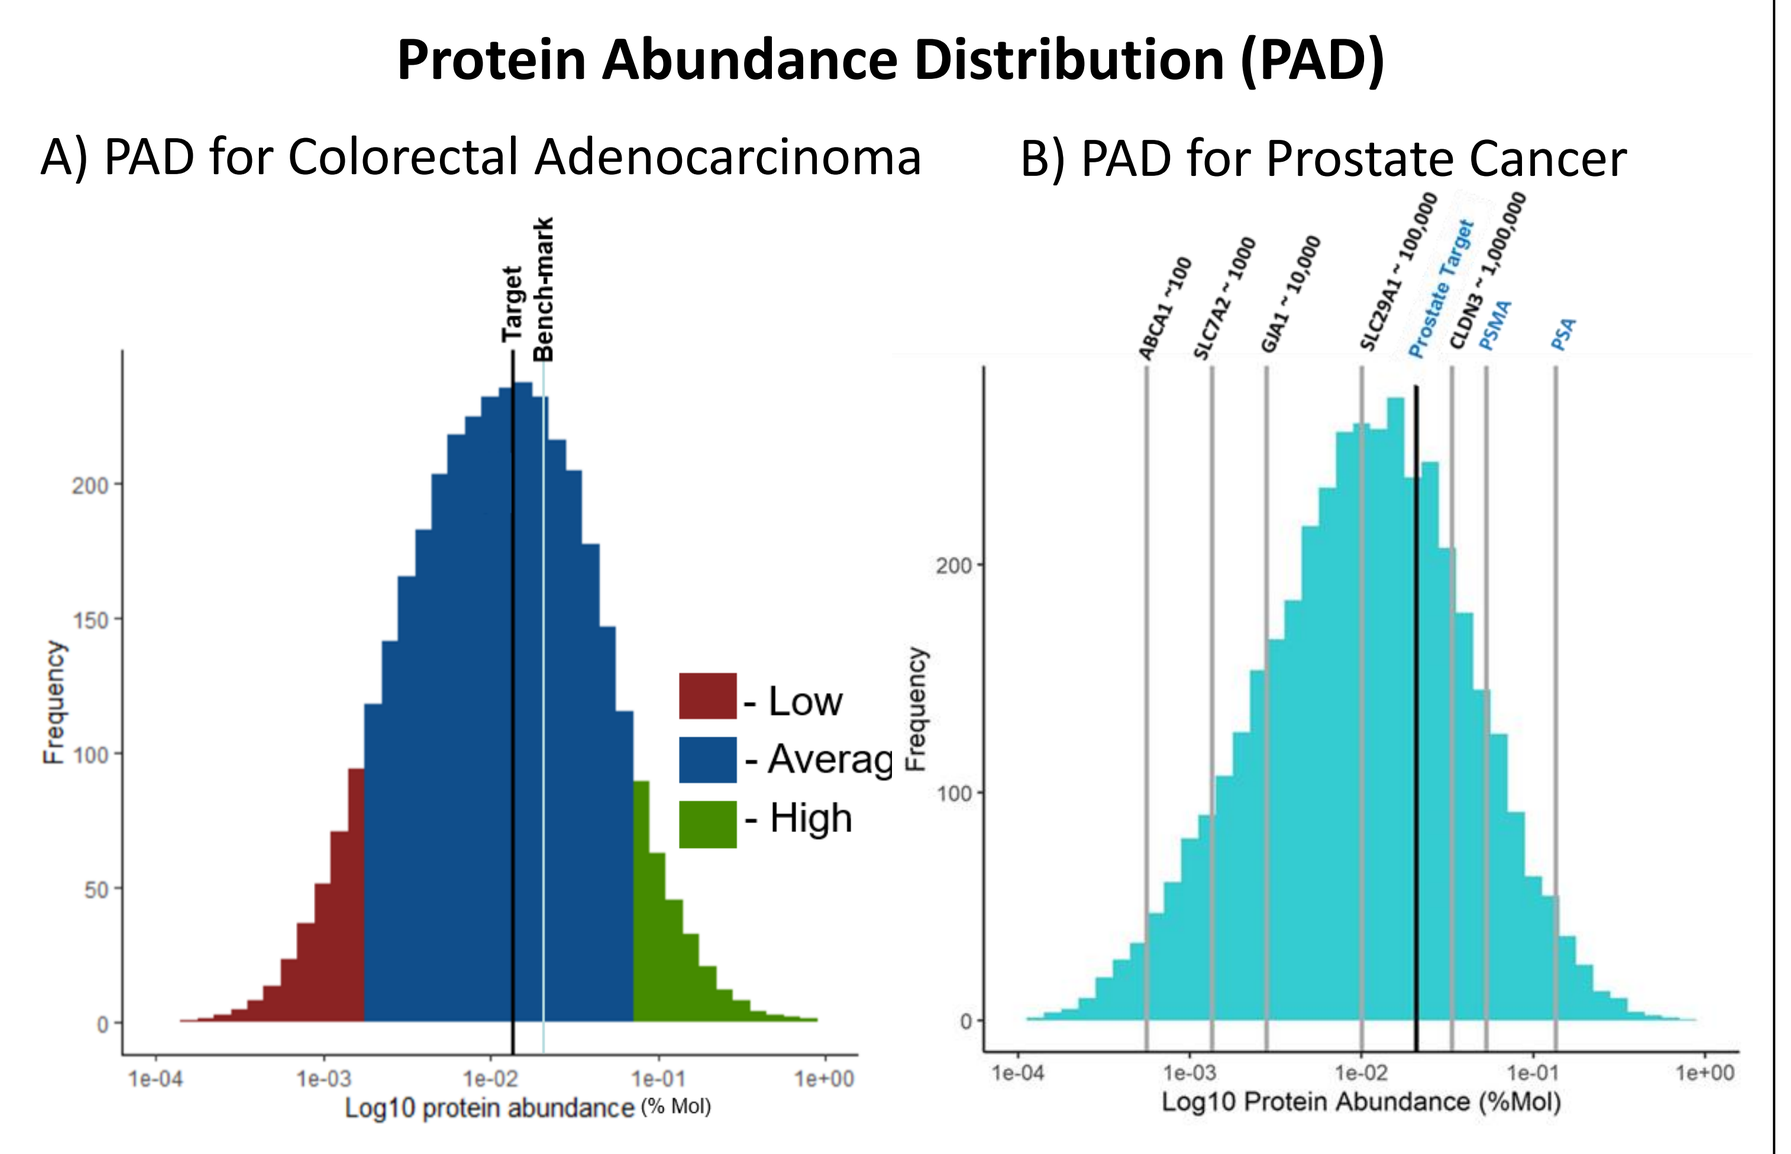

Protein Abundance Distribution (PAD)
A) PAD for Colorectal Adenocarcinoma
B) PAD for Prostate Cancer
Target
Bench-mark
- Low
- Average
- High
Frequency
Log10 protein abundance (% Mol)
Log10 Protein Abundance (%Mol)
ABCA1 ~100
SLC7A2 ~1000
GIA1 ~10,000
SLC29A1 ~100,000
Prostate Target
CLDN3 ~1,000,000
PSMA
PSA

Figure 5: Patient Abundance Distribution (PAD). Protein copies-per-cell estimated for target and a known clinical benchmark (A). Example of membrane protein expression (copies-per-cell) in patient's primary prostate tumors (B)

3.4 OGAP cancer cell line Protein Index (PI) plot is examined to see if the cell lines correlate with the cancer indications identified using the tissue PI (The OGAP cancer cell line data base has >100 cancer cell lines across multiple indications). This is also used to identify potential models as research tools.

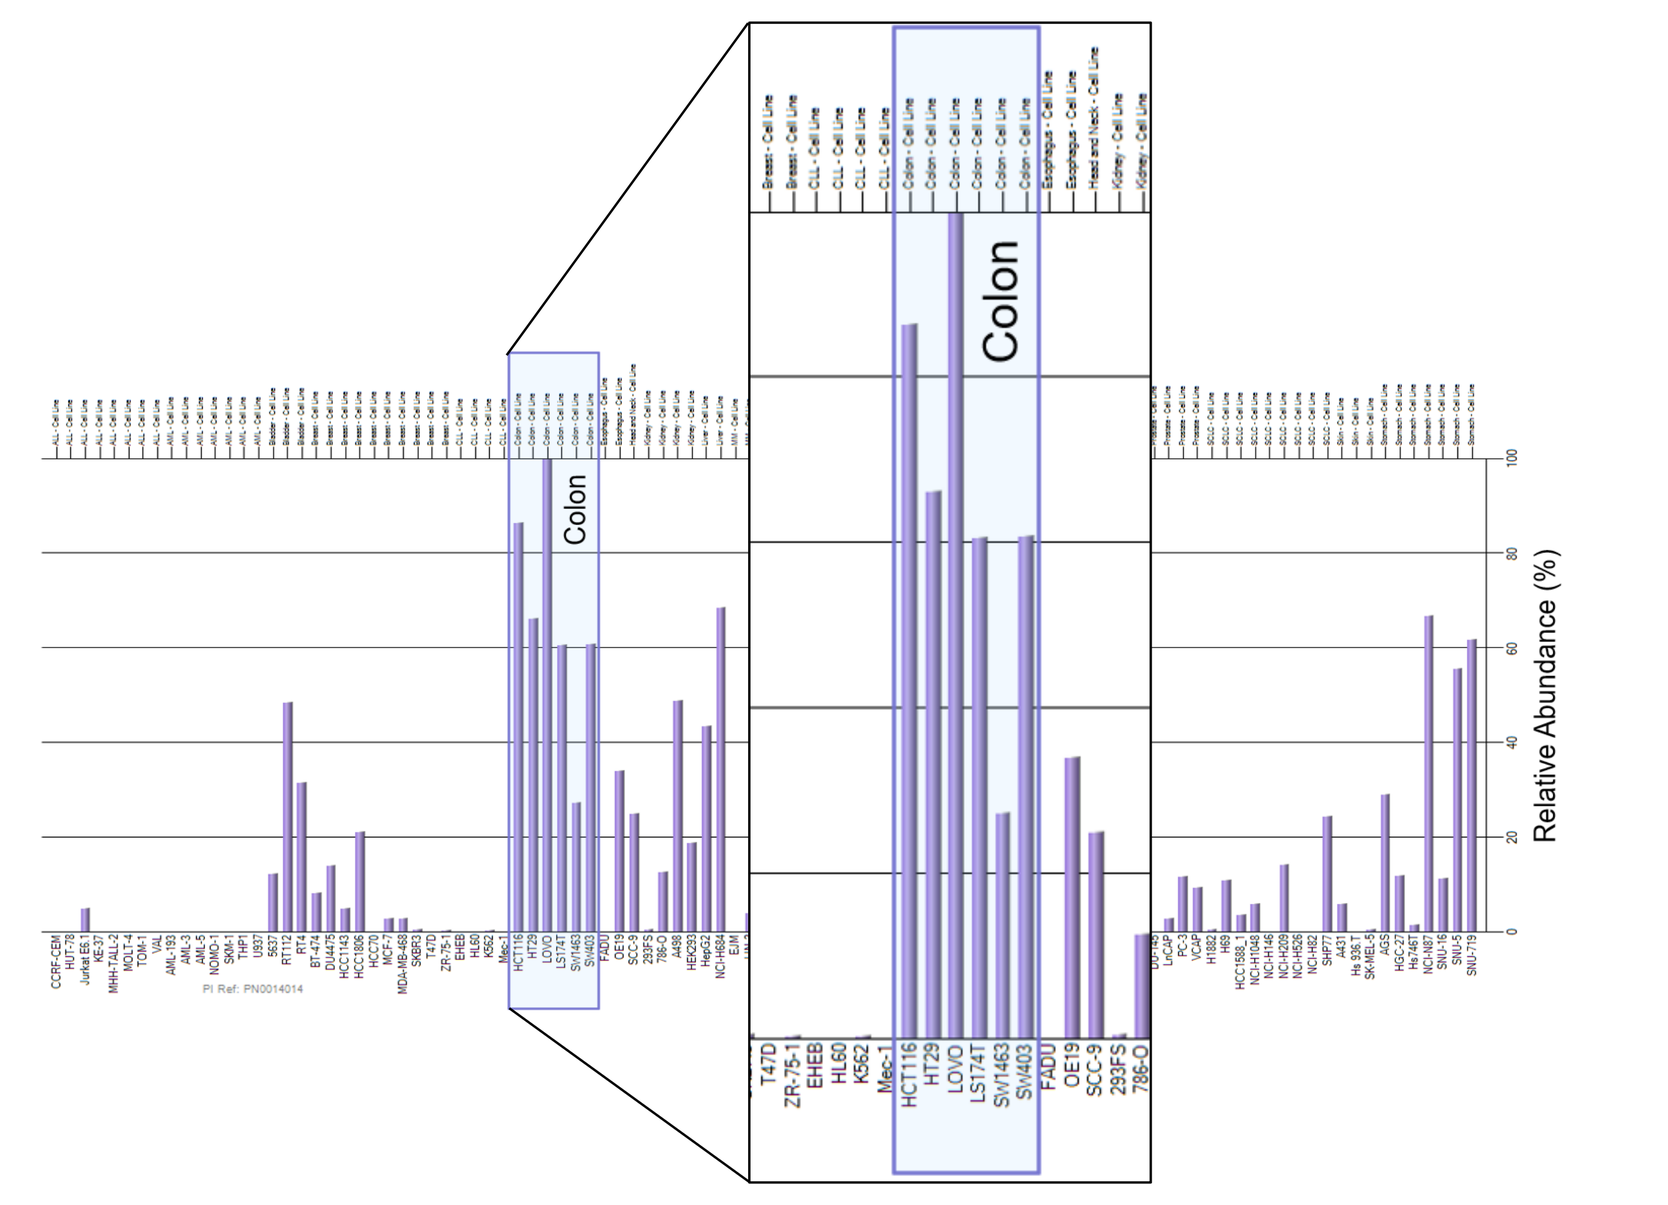

Colon
Relative Abundance (%)

Figure 6: Proteomic Cancer Cell Lines. Relative Abundance of a protein across the OGAP cancer cell line database (>100 cancer cell lines). Blue box highlights Example 1 expression in colon cancer cell lines.

3.5 Example heatmap showing membrane protein expression in different cancer indications, cell lines and NAT/normal tissues for a selection of new oncology targets. The heatmap allows rapid comparison of different targets to known clinical benchmark proteins.

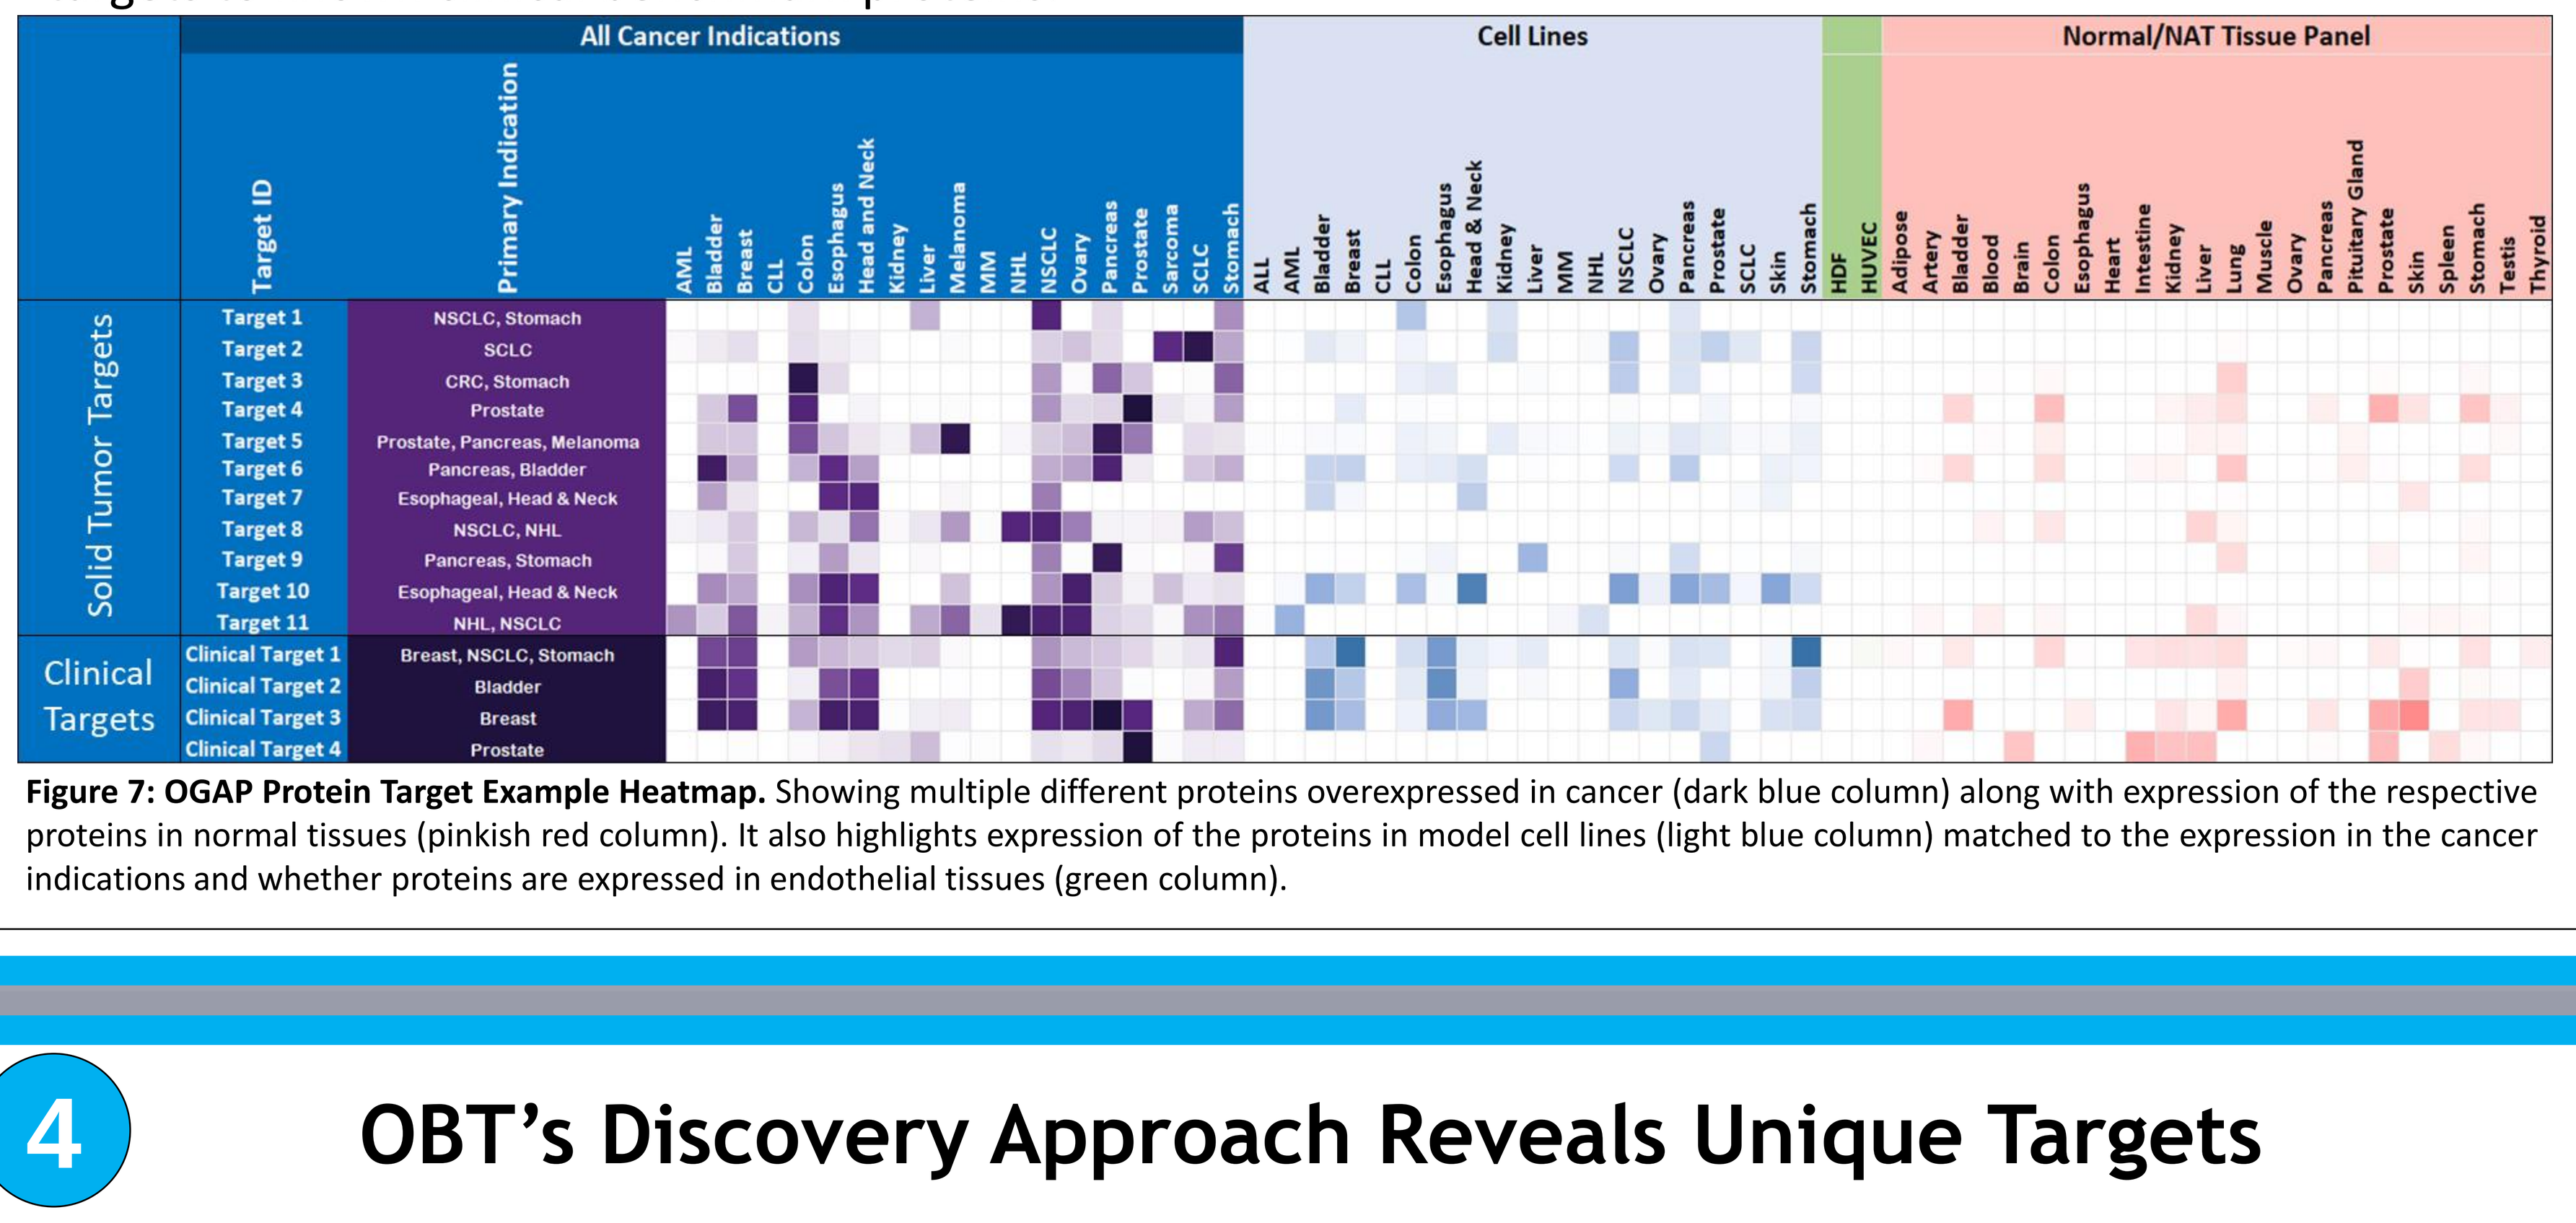

All Cancer Indications
Cell Lines
Normal/NAT Tissue Panel
Target ID
Primary Indication
Solid Tumor Targets
Clinical Targets
Target 1 NSCLC, Stomach
Target 2 SCLC
Target 3 CRC, Stomach
Target 4 Prostate
Target 5 Prostate, Pancreas, Melanoma
Target 6 Pancreas, Bladder
Target 7 Esophageal, Head & Neck
Target 8 NSCLC, NHL
Target 9 Pancreas, Stomach
Target 10 Esophageal, Head & Neck
Target 11 NHL, NSCLC
Clinical Target 1 Breast, NSCLC, Stomach
Clinical Target 2 Bladder
Clinical Target 3 Breast
Clinical Target 4 Prostate

Figure 7: OGAP Protein Target Example Heatmap. Showing multiple different proteins overexpressed in cancer (dark blue column) along with expression of the respective proteins in normal tissues (pinkish red column). It also highlights expression of the proteins in model cell lines (light blue column) matched to the expression in the cancer indications and whether proteins are expressed in endothelial tissues (green column).

4 OBT's Discovery Approach Reveals Unique Targets

- OBT has 19 years of experience of generating and analysing membrane protein expression data from over 650 cancer patient samples and cell lines (including matched primary/metastatic/NAT samples).
- OBT directly and quantitatively measures plasma membrane protein expression in patient tumors to identify novel, antibody druggable therapeutic targets, without relying on mRNA based predictions.
- Multiple, novel oncology therapeutic targets have been identified by OGAP and are now being clinically validated (3 novel OBT targets in phase I/II clinical trials).
- OBT is developing an innovative, first-in-class ADC, OBT076, which has been designed to not only target tumor cells but also overcome immune suppression in high-risk cancer patients. OBT076 was also discovered through our OGAP pipeline and developed in-house. OBT076 is currently in phase Ib clinical trials.
- We have a major strategic partnership with ImmunoGen (IMGN) to develop multiple first-in-class ADCs in solid tumors with high unmet clinical need.
- ***We are open to discuss potential discovery collaborations across multiple therapeutic antibody drug modalities.***

Reference:1. Vogel & Marcotte, Insights into the Regulation of Protein Abundance from Proteomic & Transcriptomic Analyses; Nat Rev Genet; 2013